Practical usage of eDNA in presence determination of the Allegheny woodrat (*Neotoma magister*) in southcentral Pennsylvania

Student Researcher: Camilla Groce
Mentor: Alicia Shenko

Abstract

The Allegheny woodrat (*Neotoma* magister) is a threatened species of rodent extirpated because of several factors, making it priority species in the PA State Wildlife Action Plan. This makes it important to document the presence of this species, which can be achieved through several methods of trapping. However, a new presence determination method is being explored using environmental DNA. During the summer of 2022, soil samples were collected in southcentral PA from Allegheny woodrat territory and latrine sites. DNA was extracted from the soil samples, amplified through PCR, and sent to Genewiz for analysis. Through this process, little usable DNA was found due to several factors, leading to limited DNA findings. Due to this, further testing is required and will be pursued in the future.

Introduction

The Allegheny woodrat is a threatened species of rodent from the northeastern region of the United States. It has been extirpated from its historical region due to factors such as habitat fragmentation, increased predation, spread of racoon roundworm, and loss of important food sources. This makes the Allegheny woodrat a priority species in the PA State Wildlife Action plan, which includes the maintenance of the habitat of known nest sites and encourages further research in presence surveys.

There have been many efforts towards documenting presence, mostly including physical and camera trapping, which can be considered sufficient, but are not perfect practices. This leads to a relatively unexplored method of presence determination: the use of environmental DNA. eDNA is defined by Philip Thomsen and Eske Willerslev as “genetic material obtained directly from environmental samples (soil, sediment, water, etc.) without any obvious signs of biological source material.” Compared to other methods, eDNA collection is a non-invasive practice that has high prospects for the future, especially with advances in DNA sequencing efficiency.

References

Balcom, Betsie J., and Richard H. Yahner. 1996. Microhabitat and landscape characteristics associated with the threatened allegheny woodrat. Conservation Biology 10 (2): 515-25. http://www.jstor.org/stable/2386866.

Butchkoski, Eileen. 2014. Allegheny Woodrat Species Profile. Pennsylvania Game Commission. https://www.pgc.pa.gov/Wildlife/EndangeredandThreatened/Pages/AlleghenyWoodrat.aspx.

Castleberry, S.B., Mengak, M.T. and Menken, T.E. 2014. Comparison of trapping and camera survey methods for determining presence of allegheny woodrats. Wildl. Soc. Bull., 38: 414-418. https://doi.org/10.1002/wsb.383.

Lombardi, Jason V., Steven B. Castleberry, Micheal T. Mengak, and V. K. Terrell. “Natural and Anthropogenic Factors Affecting Allegheny Woodrat Occupancy.” Wildlife Society Bulletin (2011-) 42, no. 4 (2018): 667–73. https://www.jstor.org/stable/26558996.

Ruppert, Krista M., Richard J. Kline, Md Saydur Rahman. 2019. Past, present, and future perspectives of environmental DNA (eDNA) metabarcoding: A systematic review in methods, monitoring, and applications of global eDNA. Global Ecology and Conservation, Volume 17. https://doi.org/10.1016/j.gecco.2019.e00547.

Stovall, Elizabeth Ann. 2011. Habitat characteristics and activity patterns of Allegheny woodrats within catoosa wildlife management aAea, tennessee. M.S., Tennessee Technological University, https://www.proquest.com/dissertations-theses/habitat-characteristicsactivitypatterns/docview/895084343/se-2?accountid=10488.

Thomsen, Philip F., and Eske Willerslev. 2015. Environmental DNA – An emerging tool in conservation for monitoring past and present biodiversity. Biological Conservation, Volume 183, Pages 4-18. https://doi.org/10.1016/j.biocon.2014.11.019.

Methods

Sample Collection

- 25 soil samples were collected from the Lewistown Narrows in southcentral PA, using a sanitized trowel, and contained in 50ml sterile Falcon tubes.

DNA Extraction

- Taberlet’s protocol was followed for DNA extraction. Samples were separated into 5 sections correlating to collection area. Each section received a new tube and contained 100 microliters of individual samples of the 5 original tubes according to that area—leaving each section with 500 microliters of combined sample
 - 1: included samples 1-5
 - 2: included samples 6-10
 - 3: included samples 11-15
 - 4: included samples 16-20
 - 5: included samples 21-25
- After the separation, NucleoSpin Soil Genomic DNA Extraction Kit’s protocol was followed, except for Steps 2 and 3, with the samples vortexed as described at the end of Step 3.
- After the extraction, the 5 sample containers were secured and placed in a freezer until the PCR could be performed.

PCR

- 2 PCR tests were performed. Primers LepF1, LepR1, VF1, and VR1 were mixed with the original samples (templates) and loaded into a thermal cycler. The first PCR had 35 cycles, and the goal was to amplify a segment of the COI gene.
- The PCR product after thermal cycling was mixed with loading dye and each of the 5 samples, the ladder, and two origin10al samples were loaded into the 8 wells of an agar plate. This was then placed into a gel electrophoresis machine and ran for about 20 minutes and then analyzed for DNA movement in the gel.

Lab Analysis

- The primer and template mix used in the first PCR were sent to Genewiz to be barcoded. The samples were securely packed and shipped overnight.
- The lab emailed their results and sequenced in two directions due to the quality DNA in the samples.

Results

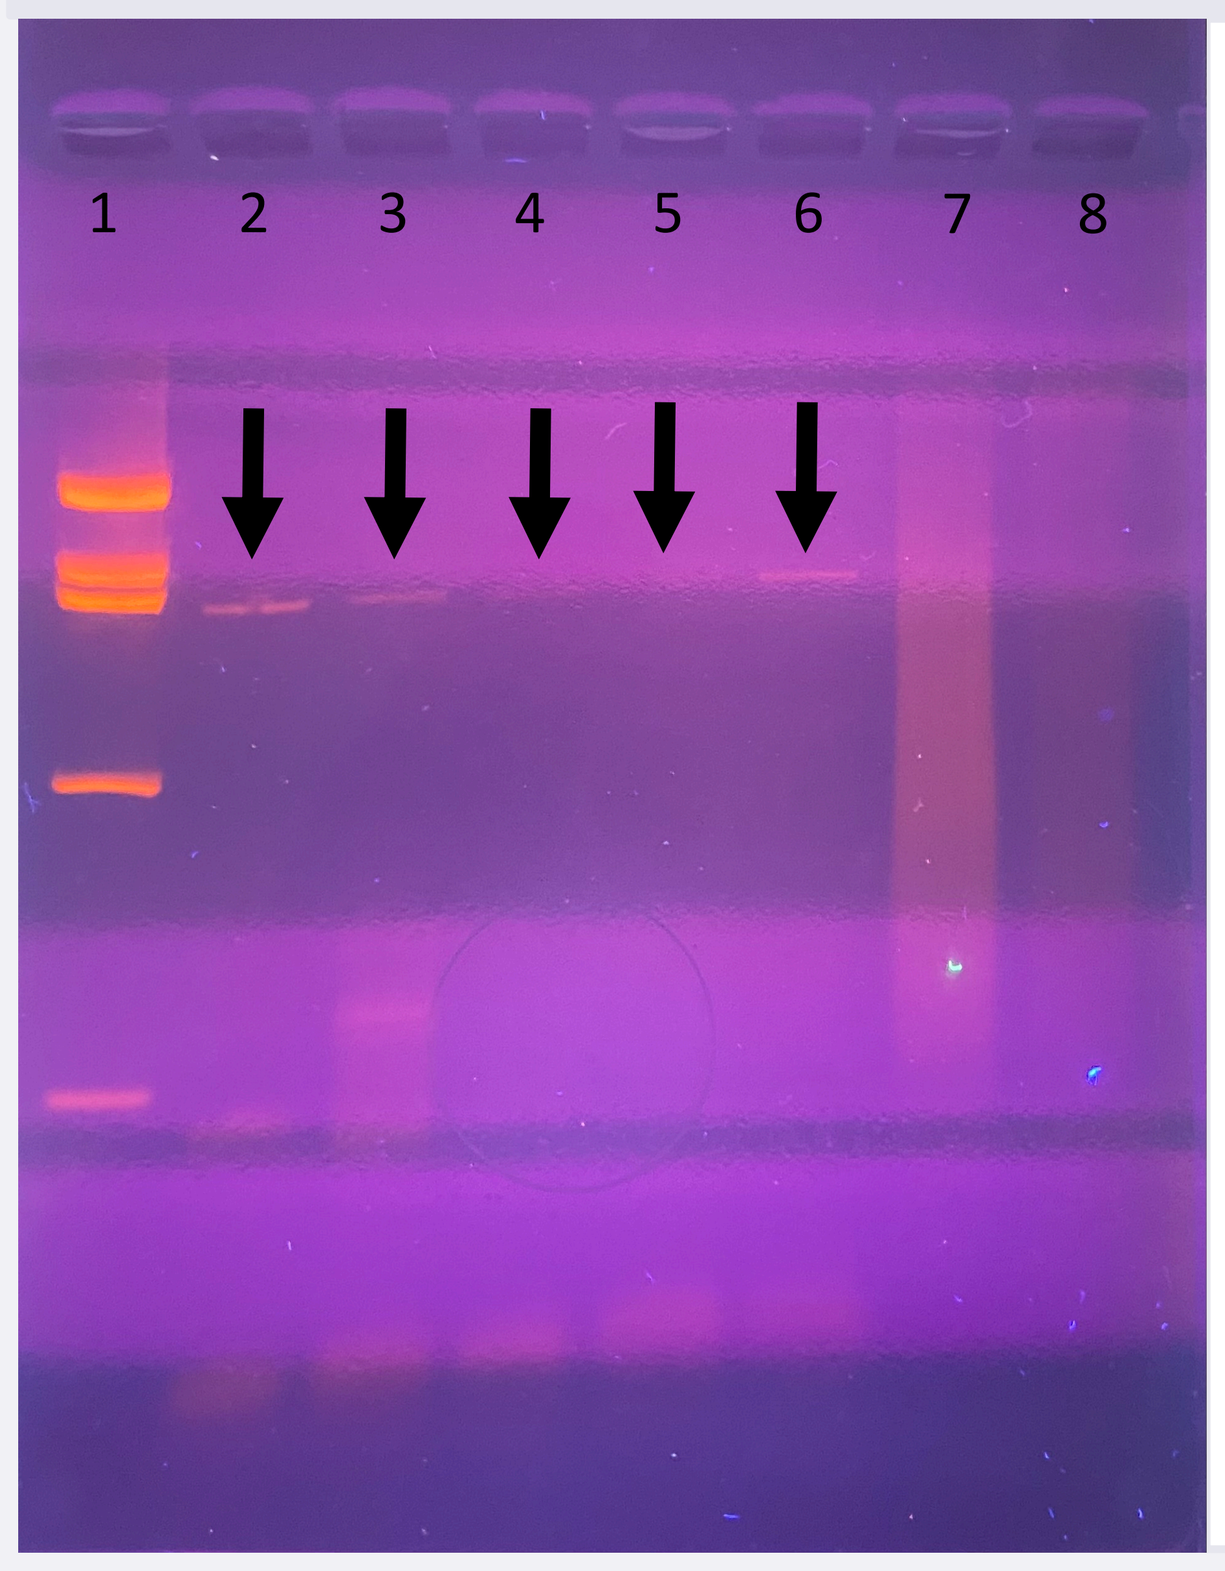
1 2 3 4 5 6 7 8

Well 1: Ladder
Well 2: Sample 1 with primers
Well 3: Sample 2 with primers
Well 4: Sample 3 with primers
Well 5: Sample 4 with primers
Well 6: Sample 5 with primers
Well 7: Sample 1 no primers
Well 8: Sample 4 no primers

Figure 1: Gel Electrophoresis of PCR Results (Above)

Figures 2-4: BLAST Results of DNA (Below)

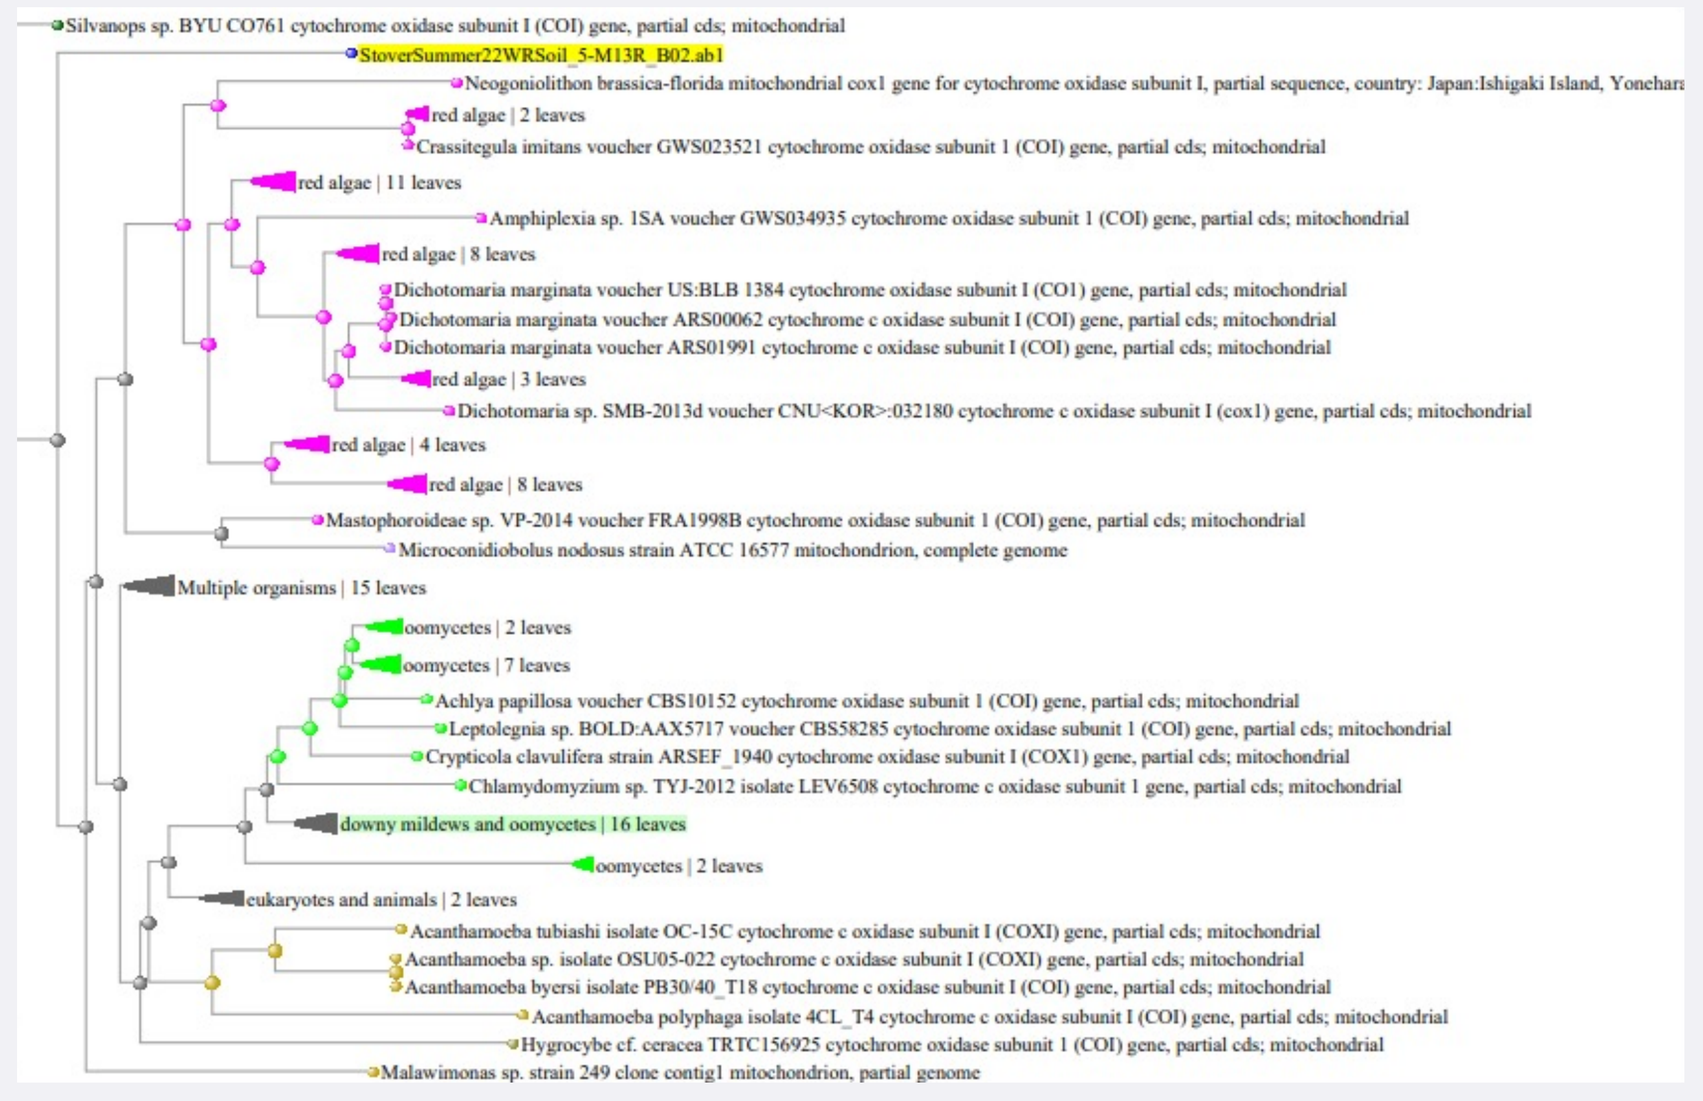

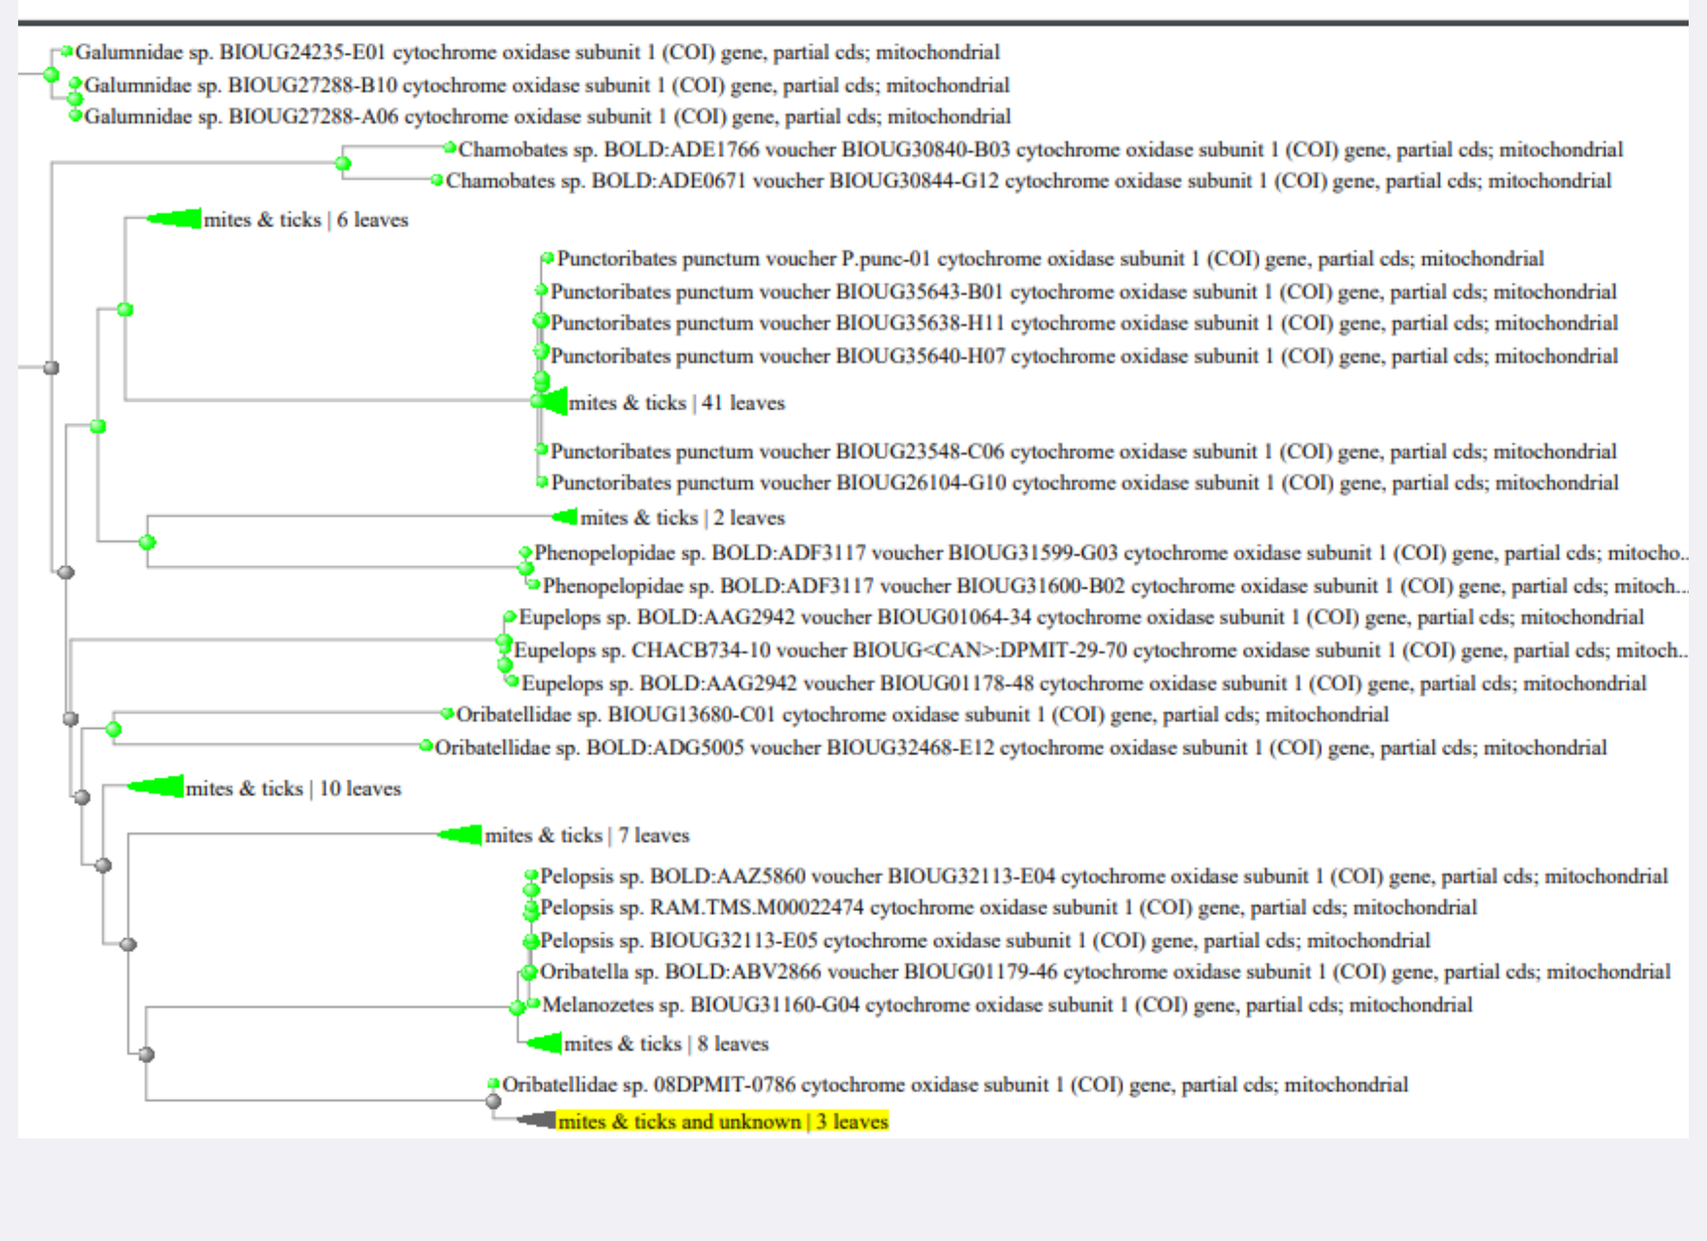

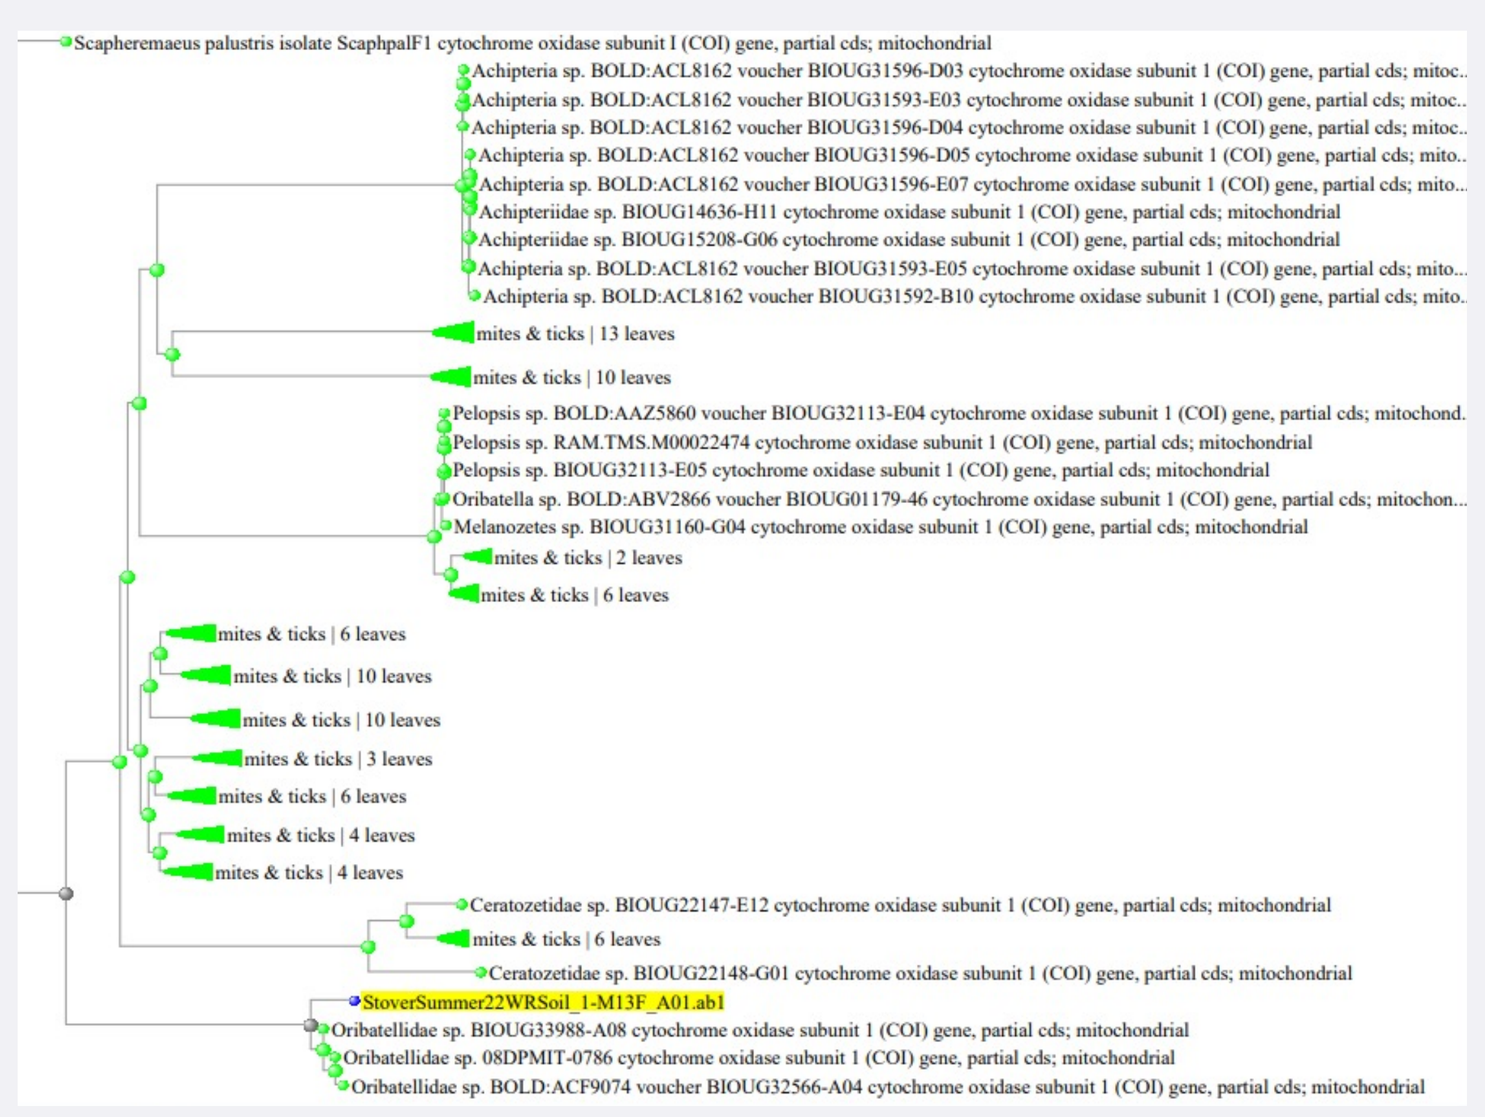

Discussion

When analyzing the PCR results on the electrophoresis gel, seen in **Figure 1**, it is notable that DNA can be found around the 800 base pair mark. This is only faintly noticeable for Samples 1 (Well 2), 2 (Well 3), and 5 (Well 6), and even less so for Samples 3 (Well 4) and 4 (Well 5). Wells 7 and 8, containing samples with no primers, showed genomic DNA, as expected with no primers being present.

Genewiz’s analysis was carried out on 2 samples overall, due to poor quality of DNA and primers. Sample 1 was sequenced in forward and reverse directions, and Sample 5 was sequenced in reverse direction. No mammalian DNA was found according to the associated BLAST search, only resulting in mite, tick, and mildew DNA, with a 97% match, as highlighted in **Figures 2-4**. The rest of the samples had no usable results.

Conclusion

With the PCR results showing only faint traces of DNA, these results were to be expected. In order to obtain more accurate DNA readings, further exploration of priming and PCR methodology is required. This includes experimenting with number of cycles in the thermal cycler, amount of template mixed with primers, and refining primer concentration. This will be performed throughout the following months to further contribute to eDNA usage knowledge, despite this specific project coming to an end.

Further research will also be dedicated to the use of eDNA to find more than just Allegheny woodrat DNA, extending to finding all mammalian DNA in the samples and documenting those species for biodiversity purposes of the areas sampled.

Acknowledgements

I would like to thank: Bristol-Myers Squibb for funding this project, the Pennsylvania Game Commission for including me in their Allegheny woodrat trapping at the Lewistown Narrows so I could gather samples, Professor Kimberly Johnston for aiding in the PCR and sequencing process, and my mentor Dr. Alicia Shenko for supporting me through this project and encouraging further research into the subject.